A phase II study of infigratinib (BGJ398) in previously-treated advanced cholangiocarcinoma containing *FGFR2* fusions

Javle M,[1] Kelley RK,[2] Roychowdhury S,[3] Weiss K-H,[4] Abou-Alfa GK,[5] Macarulla T,[6] Sadeghi S,[7] Waldschmidt D,[8] Zhu A,[9] Goyal L,[9] Borad M,[10] Yong WP,[11] Borbath I,[12] El-Khoueiry A,[13] Philip P,[14] Moran S,[15] Ye Y,[15] Ising M,[16] Lewis N,[17] Bekaii-Saab T[10]

[1]MD Anderson Cancer Center, Houston, TX, USA; [2]University of California, San Francisco, CA, USA; [3]Ohio State Comprehensive Cancer Center/James Cancer Hospital, Columbus, OH, USA; [4]University Hospital Heidelberg, Germany; [5]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [6]Hospital Vall d'Hebron, Barcelona, Spain; [7]University of California, Los Angeles, CA, USA; [8]Klinikum de Universität zu Köln, Cologne, Germany; [9]Massachusetts General Hospital, Boston, MA, USA; [10]Mayo Clinic Arizona, Scottsdale, AZ, USA; [11]National University Cancer Institute, Singapore; [12]Cliniques Universitaires St Luc Bruxelles, Brussels, Belgium; [13]USC/Kenneth Norris Comprehensive Cancer Center, Los Angeles, CA, USA; [14]Karmanos Cancer Institute, Detroit, MI, USA; [15]QED Therapeutics, San Francisco, CA, USA; [16]Novartis, Florham Park, NJ, USA; [17]Novartis Pharmaceutical Corporation, East Hanover, NJ, USA

Background

- Cholangiocarcinomas are often diagnosed at an advanced unresectable stage, with few treatment options available after disease progression while receiving gemcitabine and cisplatin first-line chemotherapy, resulting in poor patient prognosis.
- Numerous cancers have fibroblast growth factor receptor (FGFR) genomic alterations. *FGFR* translocations (i.e. fusion events) represent driver mutations in cholangiocarcinoma. They are present in 13–17% of intrahepatic cholangiocarcinomas (IHC) and may predict tumor sensitivity to FGFR inhibitors.[1–3]
- Infigratinib (BGJ398), an ATP-competitive FGFR1–3-selective oral tyrosine kinase inhibitor (Figure 1), has shown preliminary clinical activity against tumors with *FGFR* alterations.[4]
- In early-phase clinical evaluation, infigratinib showed a manageable safety profile and single-agent activity.[5,6]
- A multicenter, open-label, phase II study (NCT02150967) evaluated the antitumor activity of infigratinib in patients with previously-treated advanced IHC containing *FGFR2* fusions.

Figure 1. **Infigratinib: an oral FGFR1–3 selective kinase inhibitor**

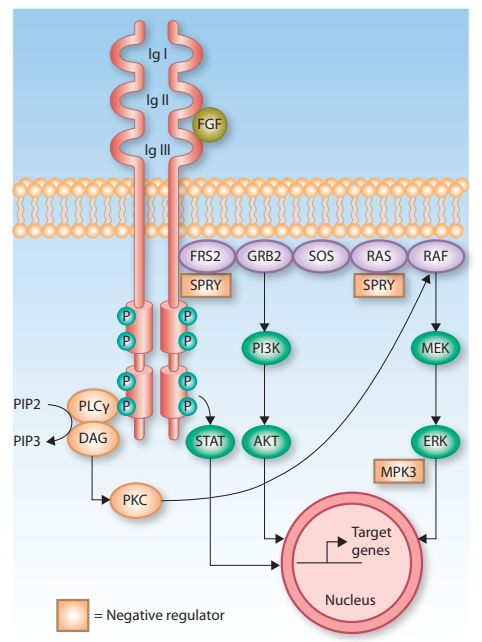
Ig I
Ig II
Ig III
FGF
FRS2
GRB2
SOS
RAS
RAF
SPRY
SPRY
PI3K
MEK
PIP2
PLCγ
PIP3
DAG
STAT
AKT
ERK
MPK3
PKC
Target genes
Nucleus
= Negative regulator

- Infigratinib is an orally-available, selective, ATP-competitive FGFR inhibitor
 – FGFR1–3 >4
- Infigratinib has proven activity in tumor models with FGFR alterations

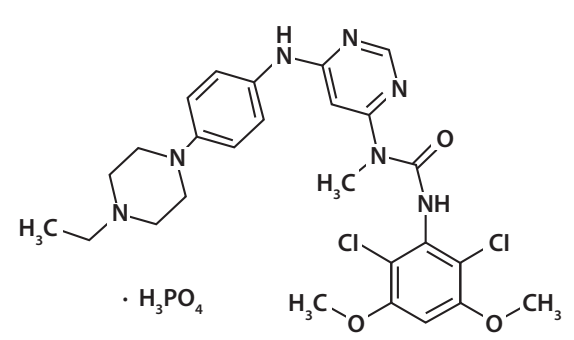
H
N
N
N
N
N
H_3C
N
O
H_3C
NH
Cl
Cl
$\cdot\ H_3PO_4$
H_3C
O
O
CH_3

Study methods

Patients

- Histologically or cytologically confirmed advanced/metastatic IHC with *FGFR2* fusions or other *FGFR* genetic alterations identified by local Clinical Laboratory Improvement Amendments – certified testing or at a central facility.
- The protocol was modified to limit enrollment to only tumors with *FGFR2* fusions.
- Measurable or evaluable disease according to RECIST (version 1.1), an ECOG performance status of 0 or 1, and evidence of disease progression after one or more prior regimens of gemcitabine-based combination therapy or gemcitabine monotherapy.

Treatment

- Patients received infigratinib 125 mg once daily for 21 days followed by 7 days off in 28-day cycles.
- To manage hyperphosphatemia, prophylactic use of sevelamer, a phosphate-binding agent, was recommended on days of infigratinib administration per the product packaging information and institutional guidelines. Patients were also instructed to adhere to a low-phosphate diet.
- Patients continued infigratinib treatment until unacceptable toxicity, disease progression, and/or investigator discretion, or consent withdrawal.
- Dose modifications were based on the worst preceding toxicity. Treatment was resumed after resolution or reduction to grade 1 toxicity, with each patient allowed two dose reductions (100 mg, 75 mg) before infigratinib discontinuation.

Outcomes

- Tumor response was assessed per RECIST version 1.1, using CT or MRI.
- Primary and secondary efficacy endpoints – see Figure 2.
- Adverse events (AEs) were assessed according to the Common Terminology Criteria for Adverse Events, version 4.03, during treatment and until 30 days after the last dose was administered.
- *FGFR* genetic alteration was required to confirm patient eligibility. These and other concurrent genetic alterations were correlated with clinical outcome.

Statistics

- Data were combined from all participating study sites for the analyses.
- Efficacy and safety analyses included all patients whose tumors had *FGFR2* fusions and received at least one infigratinib dose.

Table 1. **Baseline patient demographics and clinical characteristics**

Characteristic	N=71
Median age, years (range)	53 (28–74)
Male / female	27 (38.0) / 44 (62.0)
Race	
White	55 (77.5)
Black	3 (4.2)
Asian	4 (5.6)
Other / unknown	3 (4.2) / 6 (8.5)
ECOG performance status	
0 / 1	29 (40.8) / 42 (59.2)
Prior lines of therapy	
≤1	32 (45.1)
≥2	39 (54.9)
FGFR2 status	
Translocation positive	71 (100.0)
Mutated	5 (7.0)

Figure 2. **Open-label, phase II study design**

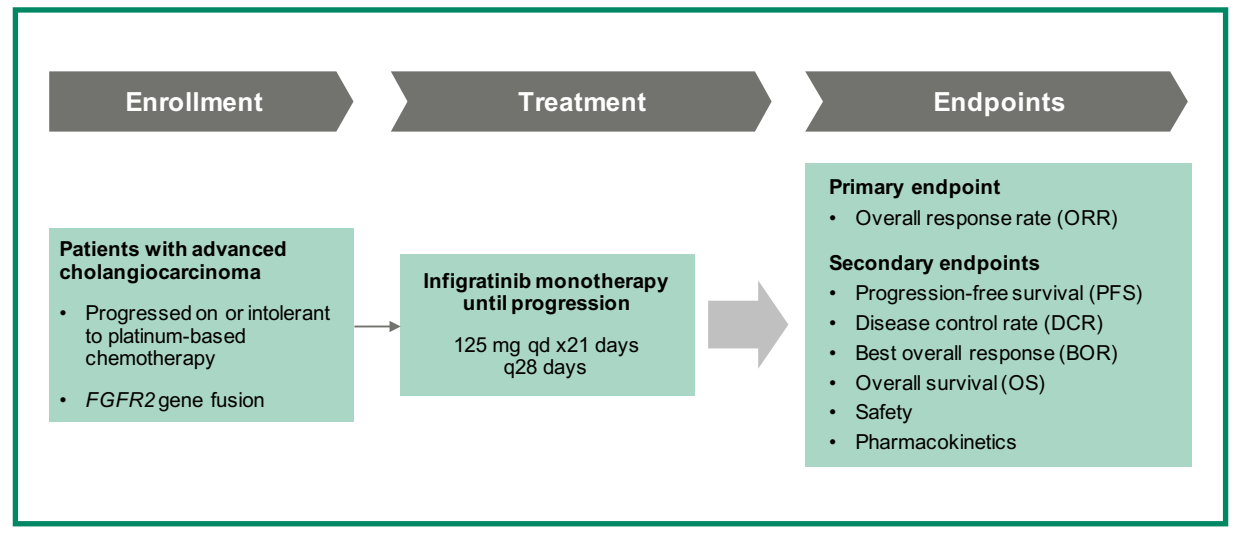
Enrollment
Treatment
Endpoints
Patients with advanced cholangiocarcinoma
• Progressed on or intolerant to platinum-based chemotherapy
• *FGFR2* gene fusion
Infigratinib monotherapy until progression
125 mg qd x21 days q28 days
Primary endpoint
• Overall response rate (ORR)
Secondary endpoints
• Progression-free survival (PFS)
• Disease control rate (DCR)
• Best overall response (BOR)
• Overall survival (OS)
• Safety
• Pharmacokinetics

Table 2. **Patient disposition**

	Number %
Total receiving treatment	71 (100.0)
Treatment ongoing	9 (12.7)
Ended treatment	62 (87.3)
Missing	1 (1.4)
Adverse event	6 (8.5)
Death	1 (1.4)
Lost to follow-up	1 (1.4)
Physician decision	5 (7.0)
Progressive disease	44 (62.0)
Subject/guardian decision	4 (5.6)

Table 3. **Clinical activity of infigratinib in advanced cholangiocarcinoma**

Efficacy outcome in all fusion patients	N=71
Overall response rate (ORR; confirmed & unconfirmed), % (95% CI)	31.0 (20.5–43.1)
Complete response, n (%)	0
Partial response – confirmed, n (%)	18 (25.4)
Stable disease, n (%)	41 (57.7)
Progressive disease, n (%)	8 (11.3)
Unknown, n (%)	4 (5.6)
Efficacy outcome in patients with potential for confirmation*	
cORR, % (95% CI)	26.9 (16.8–39.1)
cORR in patients receiving prior lines of treatment, %	
≤1 (n=28)	39.3
≥2 (n=39)	17.9
Disease control rate (DCR), % (95% CI)	83.6 (72.5–91.5)
Median duration of response, months (95% CI)	5.4 (3.7–7.4)
Median PFS, months (95% CI)	6.8 (5.3–7.6)
Median OS, months (95% CI)	12.5 (9.9–16.6)

*Patients completed (or discontinued prior to) 6 cycles. Investigator-assessed.

Figure 3. **Efficacy of infigratinib in *FGFR2* fusion-positive cholangiocarcinoma**

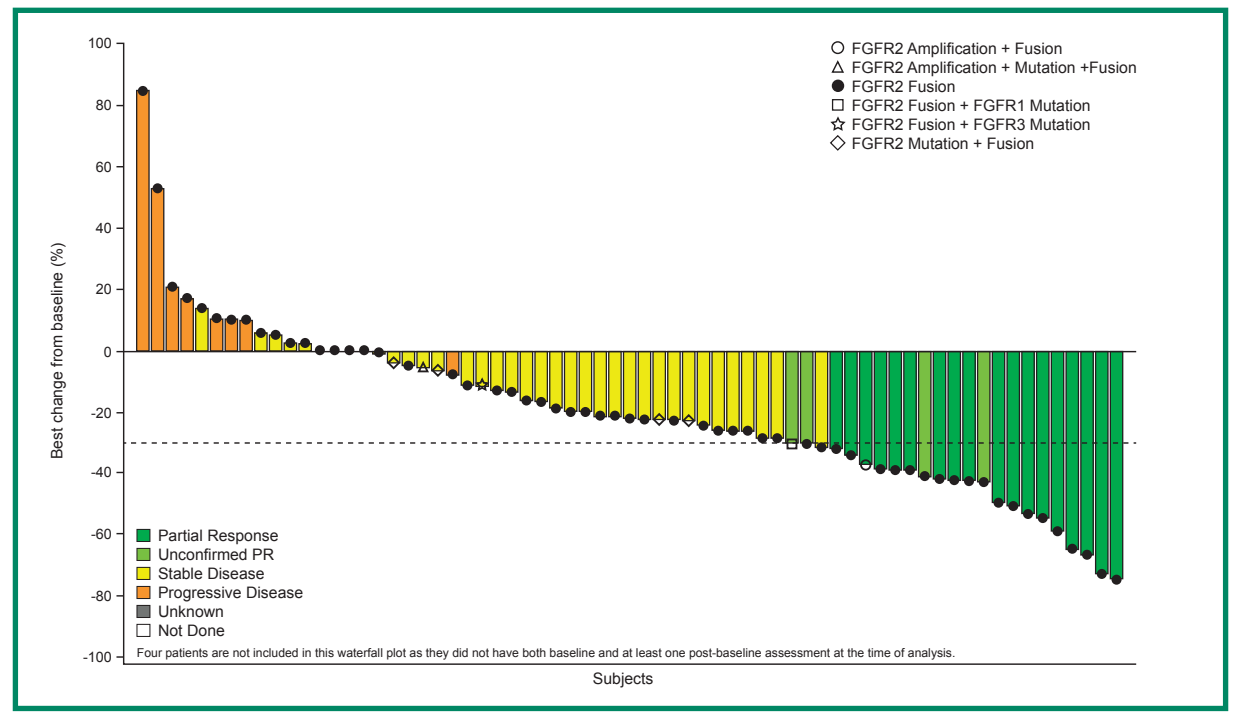
Best change from baseline (%)
100
80
60
40
20
0
-20
-40
-60
-80
-100
FGFR2 Amplification + Fusion
FGFR2 Amplification + Mutation +Fusion
FGFR2 Fusion
FGFR2 Fusion + FGFR1 Mutation
FGFR2 Fusion + FGFR3 Mutation
FGFR2 Mutation + Fusion
Partial Response
Unconfirmed PR
Stable Disease
Progressive Disease
Unknown
Not Done
Four patients are not included in this waterfall plot as they did not have both baseline and at least one post-baseline assessment at the time of analysis.
Subjects

Figure 4. **Tumor response with treatment exposure**

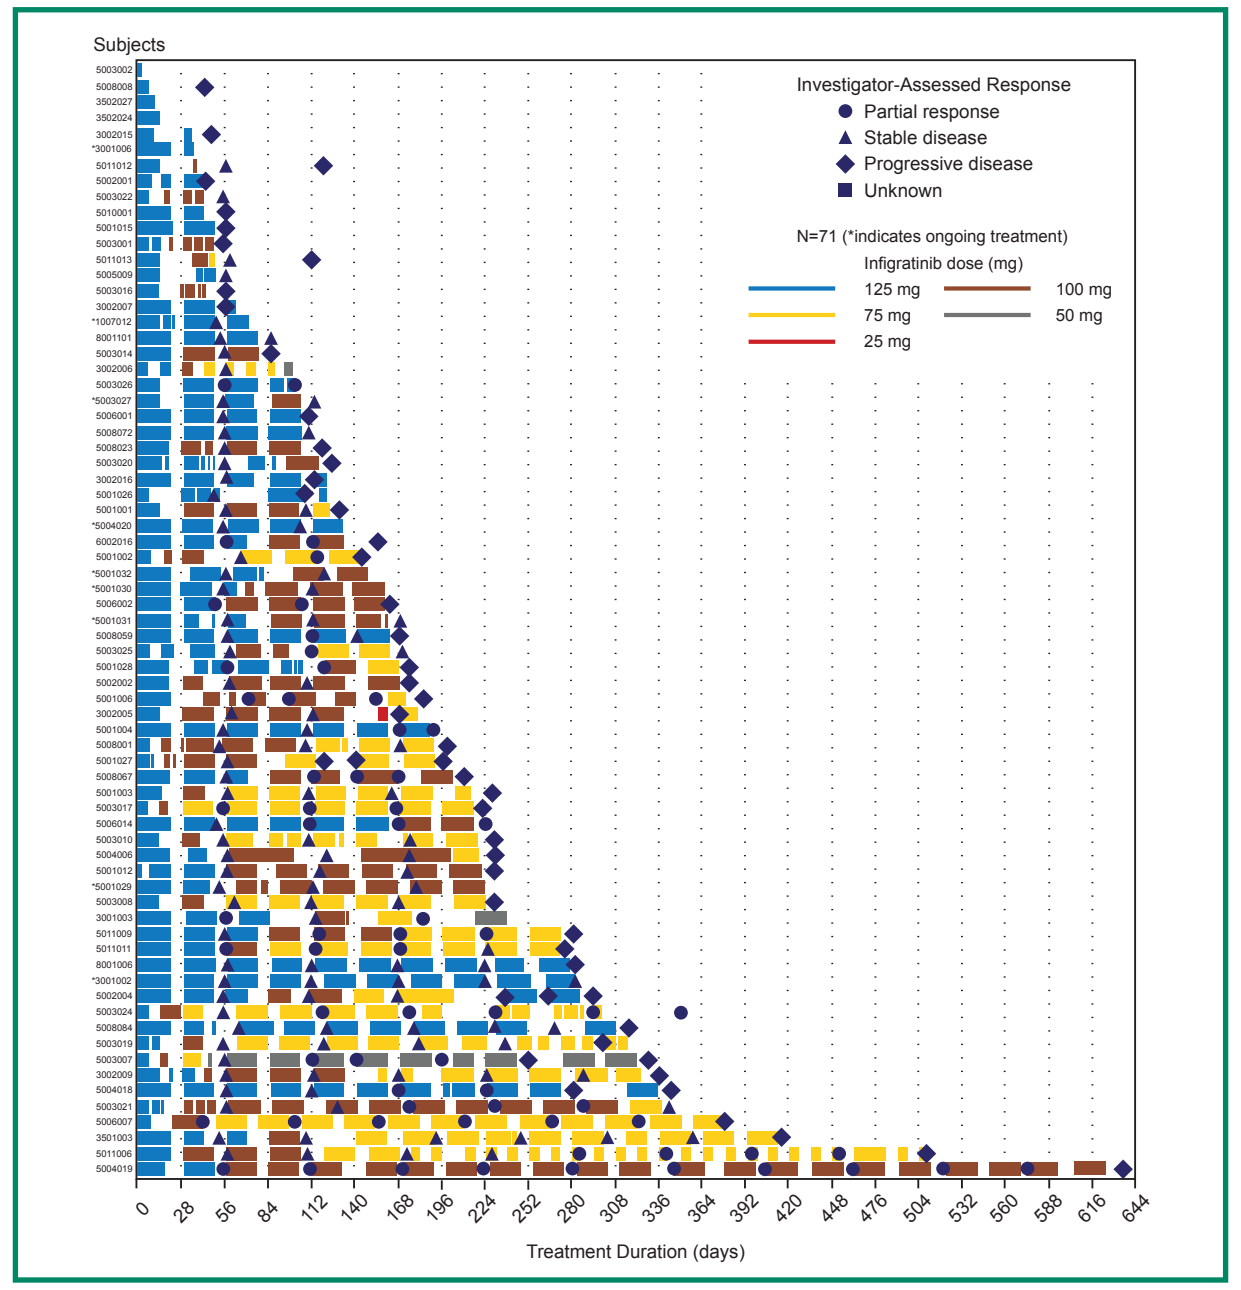
Subjects
Investigator-Assessed Response
Partial response
Stable disease
Progressive disease
Unknown
N=71 (*indicates ongoing treatment)
Infigratinib dose (mg)
125 mg
100 mg
75 mg
50 mg
25 mg
0
28
56
84
112
140
168
196
224
252
280
308
336
364
392
420
448
476
504
532
560
588
616
644
Treatment Duration (days)

Response to infigratinib in *FGFR2* fusion-positive cholangiocarcinoma

- 50-year old male with intrahepatic cholangiocarcinoma.
- s/p right hepatectomy, systemic chemotherapy with gemcitabine and cisplatin and pembrolizumab.
- Partial response on infigratinib noted at first restaging in multiple liver metastases.
- Molecular profile: *FGFR2* rearrangement, PTCH1, ARID1A, BCORL1, MAP2K4, MLL3, NUP93, SPEN, TP53. MSI high and TMB-high.

Before

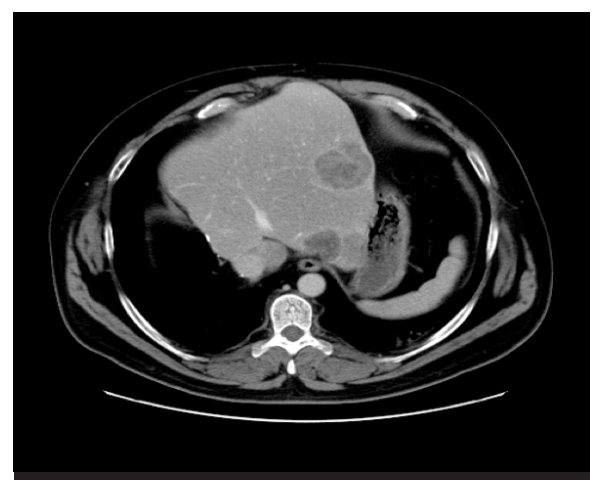

After

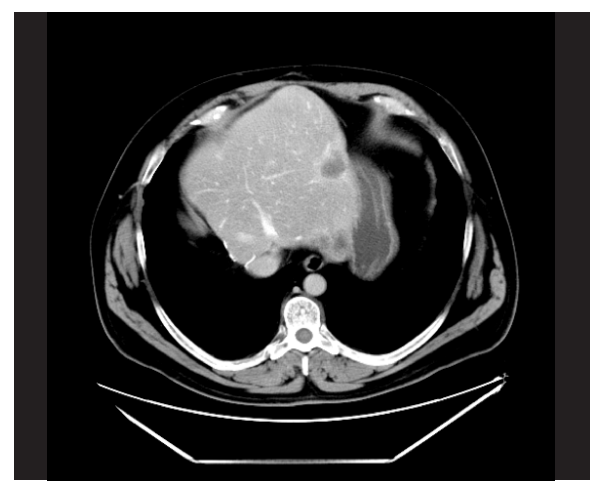

Table 4. **Infigratinib safety profile: any grade AEs ≥20%**

Number of patients (%)	Any grade	Grade 3/4
Hyperphosphatemia	52 (73.2)	9 (12.7)
Fatigue	35 (49.3)	3 (4.2)
Stomatitis	32 (45.1)	7 (9.9)
Alopecia	27 (38.0)	0
Constipation	25 (35.2)	1 (1.4)
Dry eye	23 (32.4)	0
Dysgeusia	23 (32.4)	0
Arthralgia	21 (29.6)	1 (1.4)
Palmar-plantar erythrodysesthesia syndrome	19 (26.8)	4 (5.6)
Dry mouth	18 (25.4)	0
Dry skin	18 (25.4)	0
Diarrhea	17 (23.9)	2 (2.8)
Hypophosphatemia	17 (23.9)	10 (14.1)
Nausea	17 (23.9)	1 (1.4)
Vomiting	17 (23.9)	1 (1.4)
Hypercalcemia	16 (22.5)	3 (4.2)
Vision blurred	16 (22.5)	0
Decreased appetite	15 (21.1)	1 (1.4)
Weight decreased	15 (21.1)	2 (2.8)

Table 5. **Infigratinib safety profile: grade 3/4 AEs >3%**

Number of patients (%)	Grade 3/4
Hypophosphatemia	10 (14.1)
Hyperphosphatemia	9 (12.7)
Hyponatremia	8 (11.3)
Stomatitis	7 (9.9)
Lipase increased	4 (5.6)
Palmar-plantar erythrodysesthesia syndrome	4 (5.6)
Abdominal pain	3 (4.2)
Anemia	3 (4.2)
Blood alkaline phosphatase increased	3 (4.2)
Fatigue	3 (4.2)
Hypercalcemia	3 (4.2)

Conclusions

- Infigratinib is an oral, FGFR1—3-selective TKI that shows meaningful clinical activity against chemotherapy-refractory cholangiocarcinoma containing *FGFR2* fusions.
- Infigratinib-associated toxicity is manageable with phosphate binders and routine supportive care.
- This promising antitumor activity and manageable safety profile supports continued development of infigratinib in this highly selected patient population.

Acknowledgements

The authors would like to acknowledge the following:

- CBGJ398X2204 study investigators and participating patients.
- Jeff Cai for programming and Ai Li for review of content (QED Therapeutics).
- Lee Miller (Miller Medical Communications) for provision of medical writing support for this poster. This work was funded by QED Therapeutics.

References

1. Graham RP, et al. Fibroblast growth factor receptor 2 translocations in intrahepatic cholangiocarcinoma. Hum Pathol 2014;45:1630–8.
2. Ross JS, et al. New routes to targeted therapy of intrahepatic cholangiocarcinomas revealed by next-generation sequencing. Oncologist 2014;19:235–42.
3. Farshidfar F, et al. Integrative genomic analysis of cholangiocarcinoma identifies distinct IDH-mutant molecular profiles. Cell Rep 2017;18:2780–94.
4. Guagnano V, et al. FGFR genetic alterations predict for sensitivity to NVP-BGJ398, a selective pan-FGFR inhibitor. Cancer Discov 2012;2:1118–33.
5. Nogova L, et al. Evaluation of BGJ398, a fibroblast growth factor receptor 1-3 kinase inhibitor, in patients with advanced solid tumors harboring genetic alterations in fibroblast growth factor receptors: Results of a global phase I, dose-escalation and dose-expansion study. J Clin Oncol 2017;35:157–65.
6. Javle MM, et al. A phase 2 study of BGJ398 in patients (pts) with advanced or metastatic FGFR-altered cholangiocarcinoma (CCA) who failed or are intolerant to platinum-based chemotherapy. J Clin Oncol 2016;34(suppl 4S; abstr 335).
7. Helsten et al. The FGFR landscape in cancer: analysis of 4,853 tumors by next-generation sequencing. Clin Cancer Res 2016;22:259–67.

Presented at the Cholangiocarcinoma Foundation Annual Conference 2019, 30th January – 2nd February, Salt Lake City, Utah, USA.

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.